PHARMACOKINETICS MODELING OF GLUCAGON AND A NOVEL GLUCAGON ANALOGUE AFTER SUBCUTANEOUS ADMINISTRATION IN DOGS

Sabrina Lyngbye Wendt[1,2], Anders Valeur[1], Henrik Madsen[2], John Bagterp Jørgensen[2], Carsten Boye Knudsen[1]

[1] Zealand Pharma A/S, [2] Technical University of Denmark

Background

Currently available hypoglycemic glucagon rescue kits are difficult to handle and need reconstitution immediately before use due to the instability of native glucagon in solution. A novel Zealand Pharma invented glucagon analogue (ZP-GA-1) with increased stability in liquid formulation has potential for application in a ready-to-use rescue pen. Pharmacokinetic (PK) characteristics similar to native glucagon and fast on-set of action are critical for success.

Methods

Five dogs were included in a randomized cross-over study. At four dosing occasions each dog received a subcutaneous (SC) bolus injection of 20 or 120 nmol/kg (D) native glucagon or ZP-GA-1. Blood samples were collected at 0, 5, 10, 15, 20, 30, 40, 50, 60, 75, 110, 140, and 180 minutes after dose administration. Sample concentration (y) of glucagon were analyzed using the MSD immuno assay. Plasma concentration (y) of ZP-GA-1 were analyzed using LC-MS/MS.

A one compartment model with extravascular bolus administration was fitted to each individual dataset and each population by minimizing the y^{-1}-weighted residual sum of squares using a BFGS quasi-Newton optimization algorithm in R version 3.1.0 “Spring Dance”. T_{max}, a surrogate marker of on-set of action, and C_{max} was obtained from the fit.

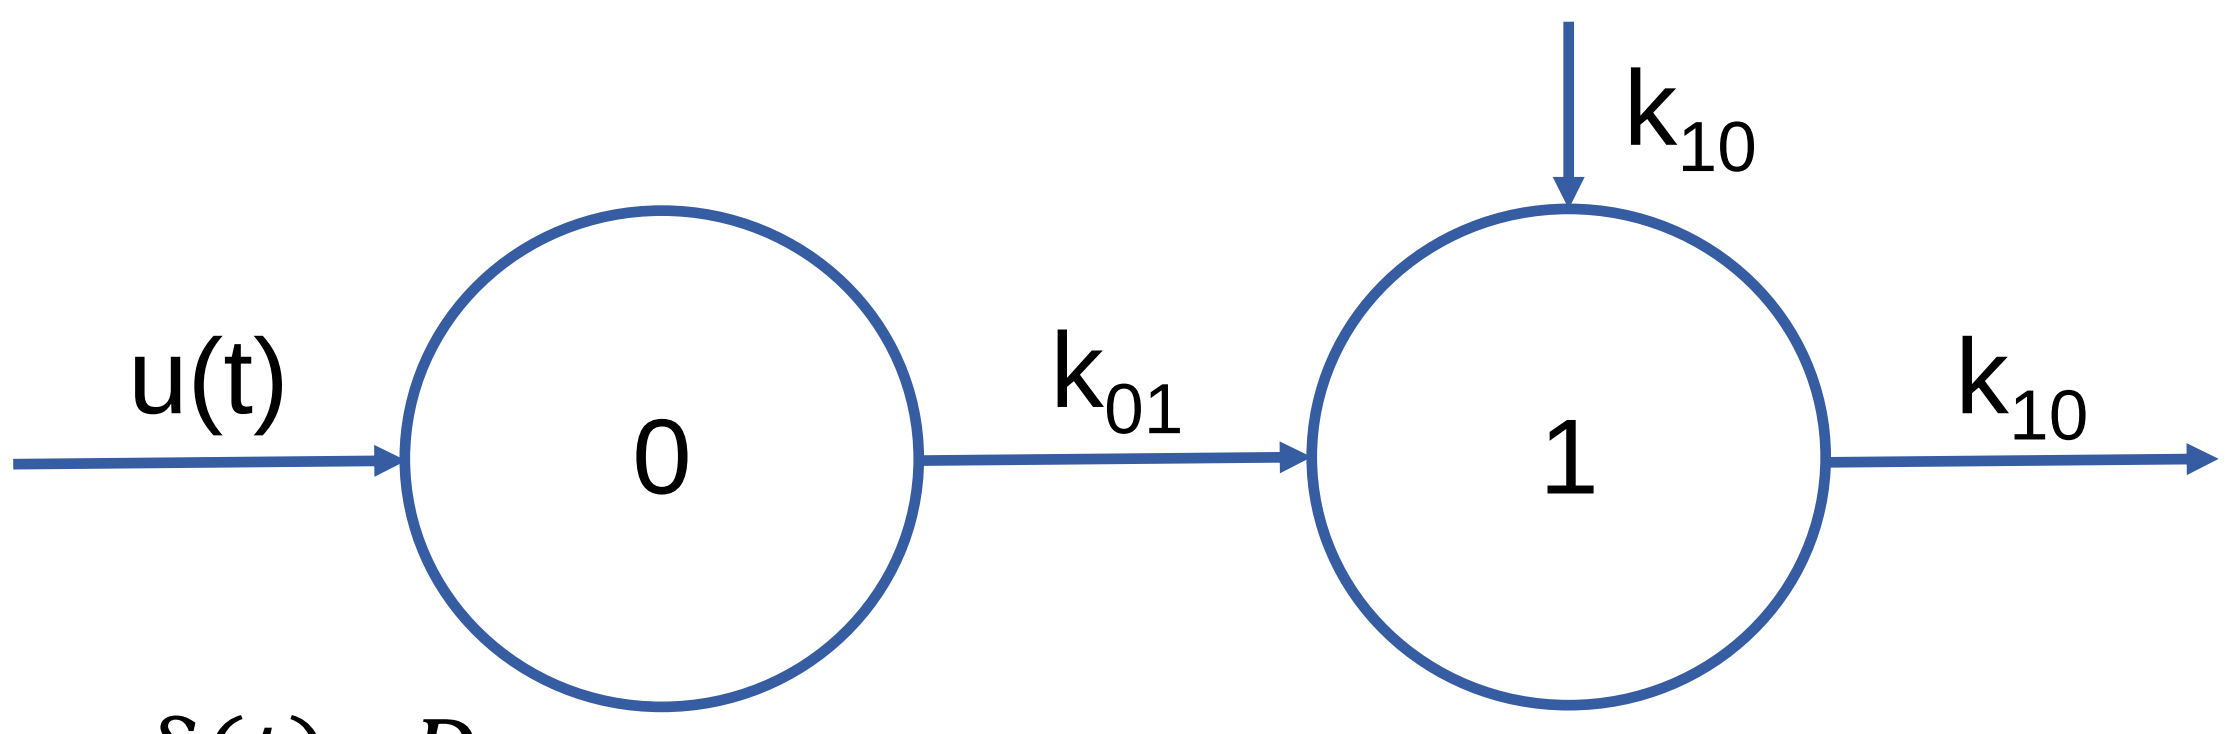
k10
u(t)
0
k01
1
k10

$$u(t) = \delta(t) \cdot D$$

$$\dot{x}_0(t) = u(t) - k_{01} \cdot x_0(t) \qquad x_0(0) = 0$$

$$\dot{x}_1(t) = k_{01} \cdot \frac{x_0(t)}{V} - k_{10} \cdot (x_1(t) - C_0) \qquad x_1(0) = C_0$$

Results

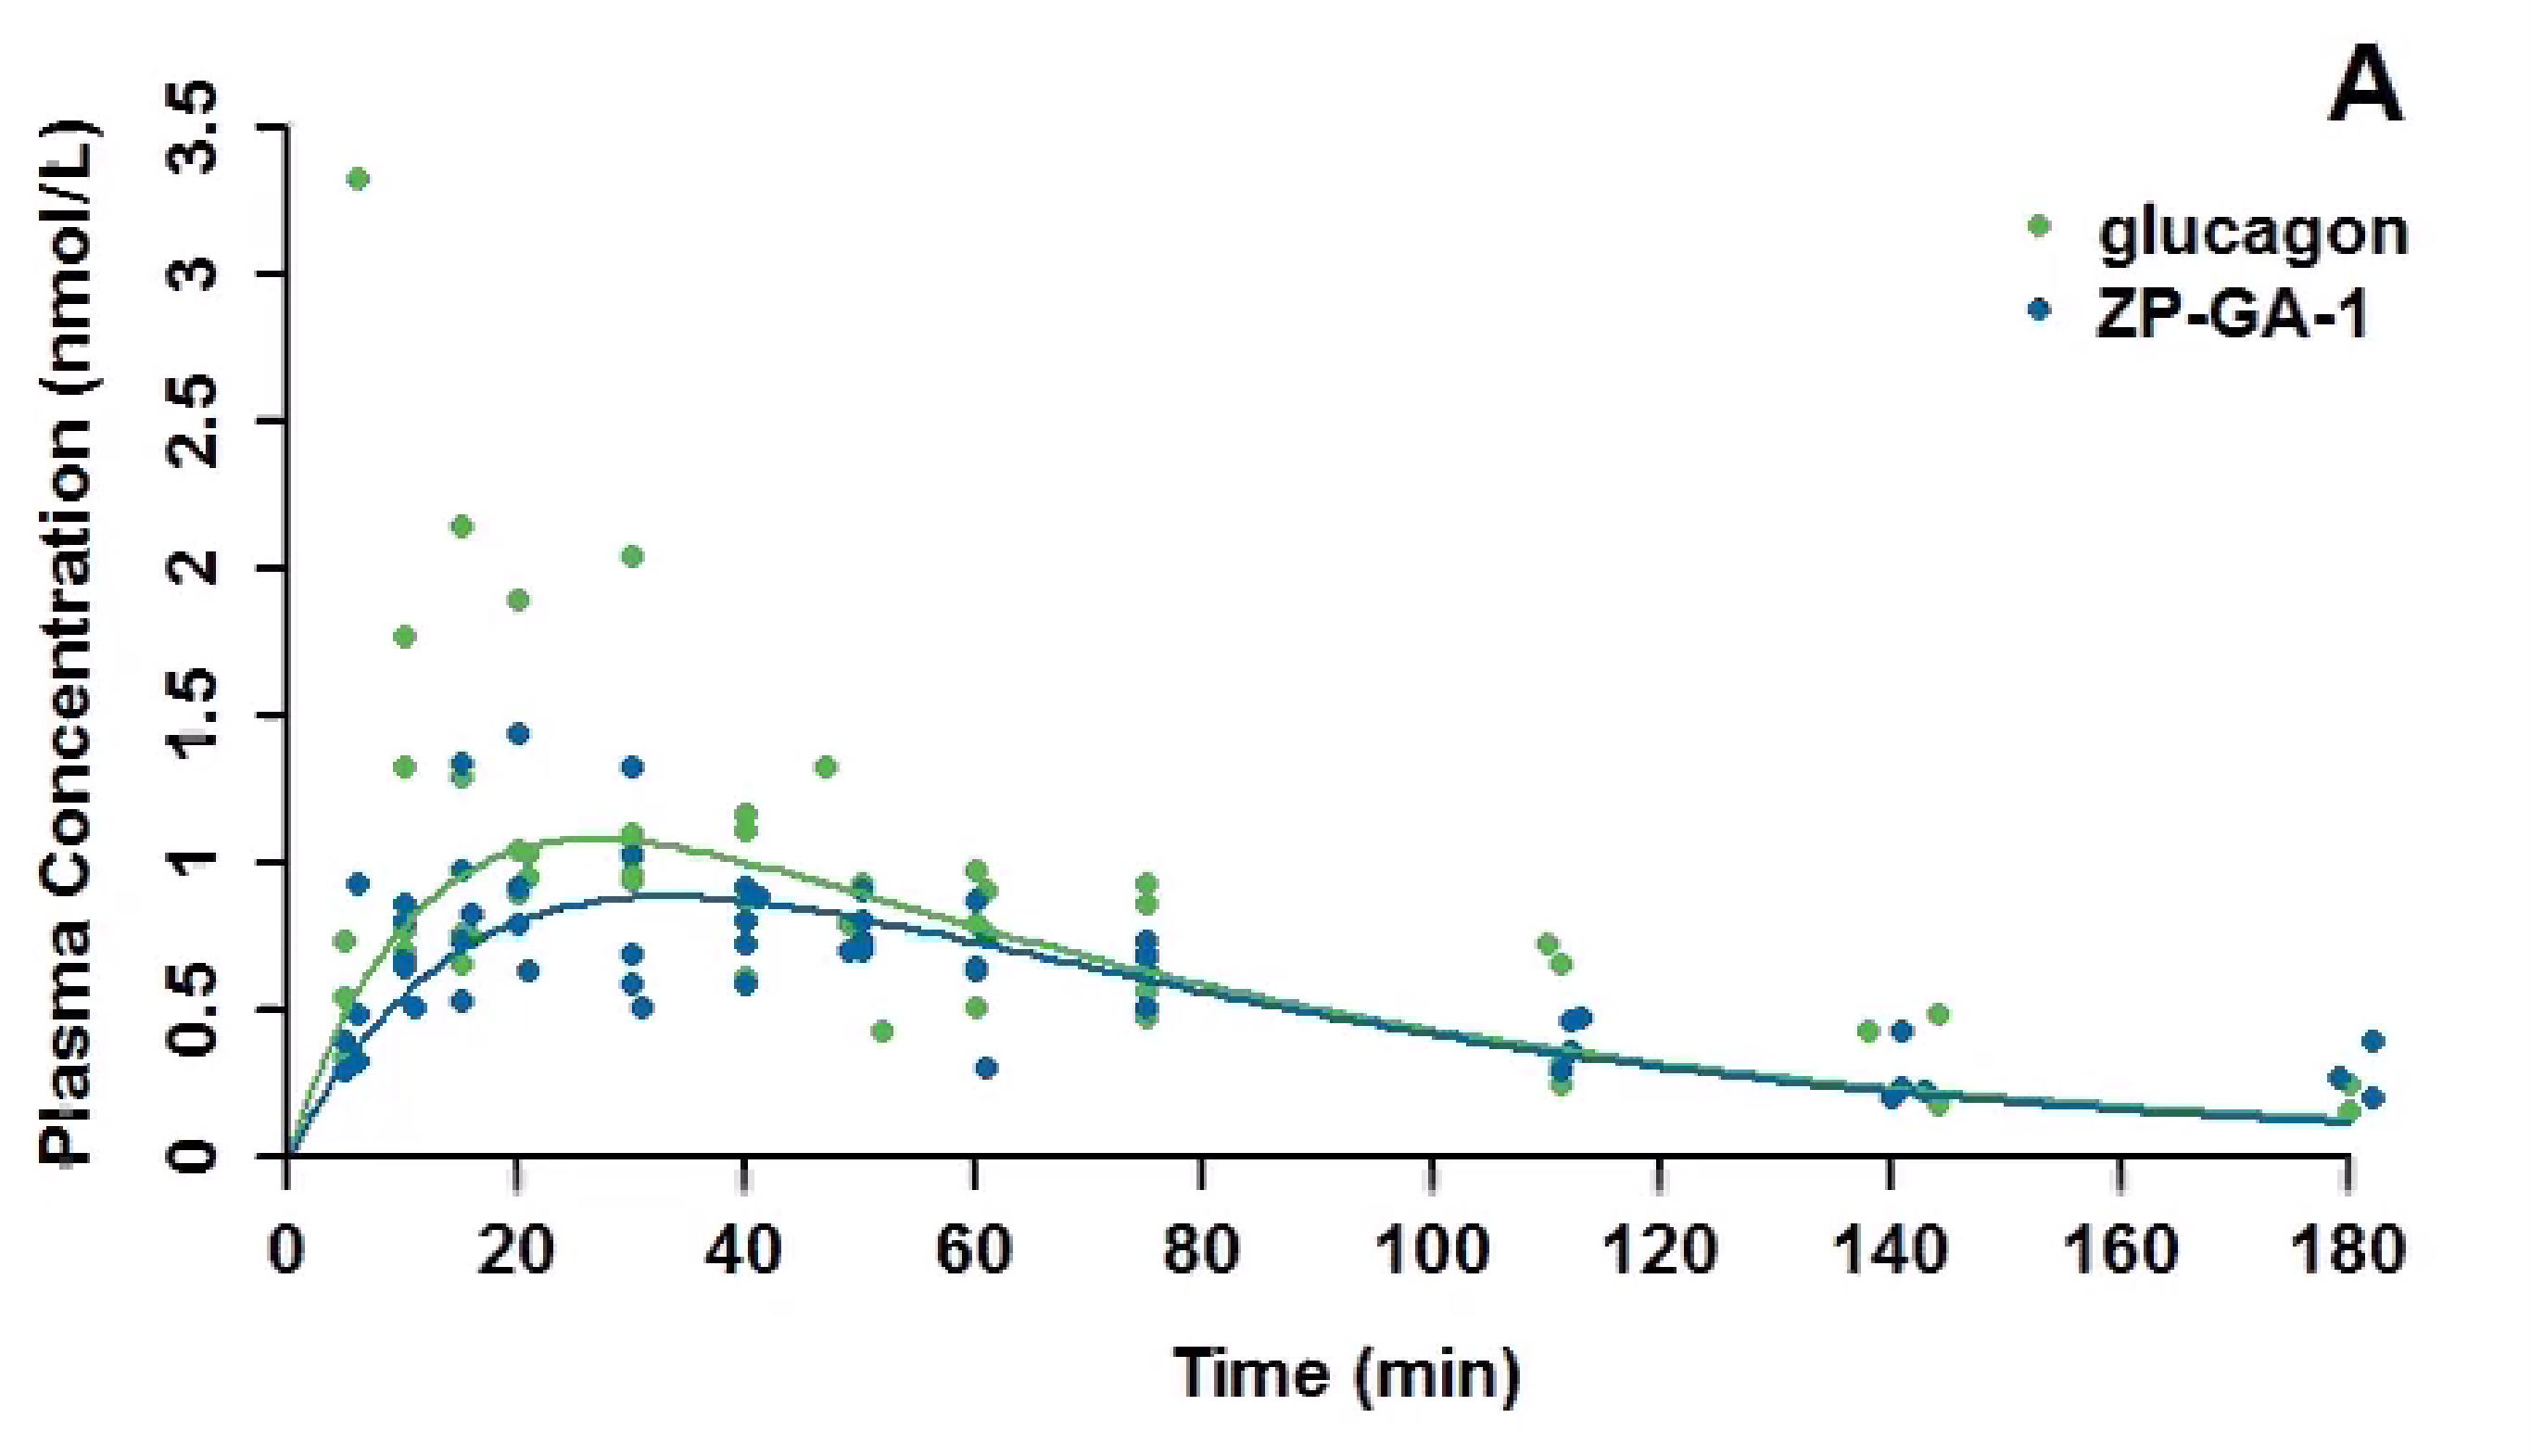
A
glucagon
ZP-GA-1
Plasma Concentration (nmol/L)
0
0.5
1
1.5
2
2.5
3
3.5
0
20
40
60
80
100
120
140
160
180
Time (min)

Figure 1: PK profiles after low SC dose. A) Raw observations with best fit to population data. B) Semi logarithmic plot with observations and best fit to population data. C) Semi logarithmic plot with individual fits.

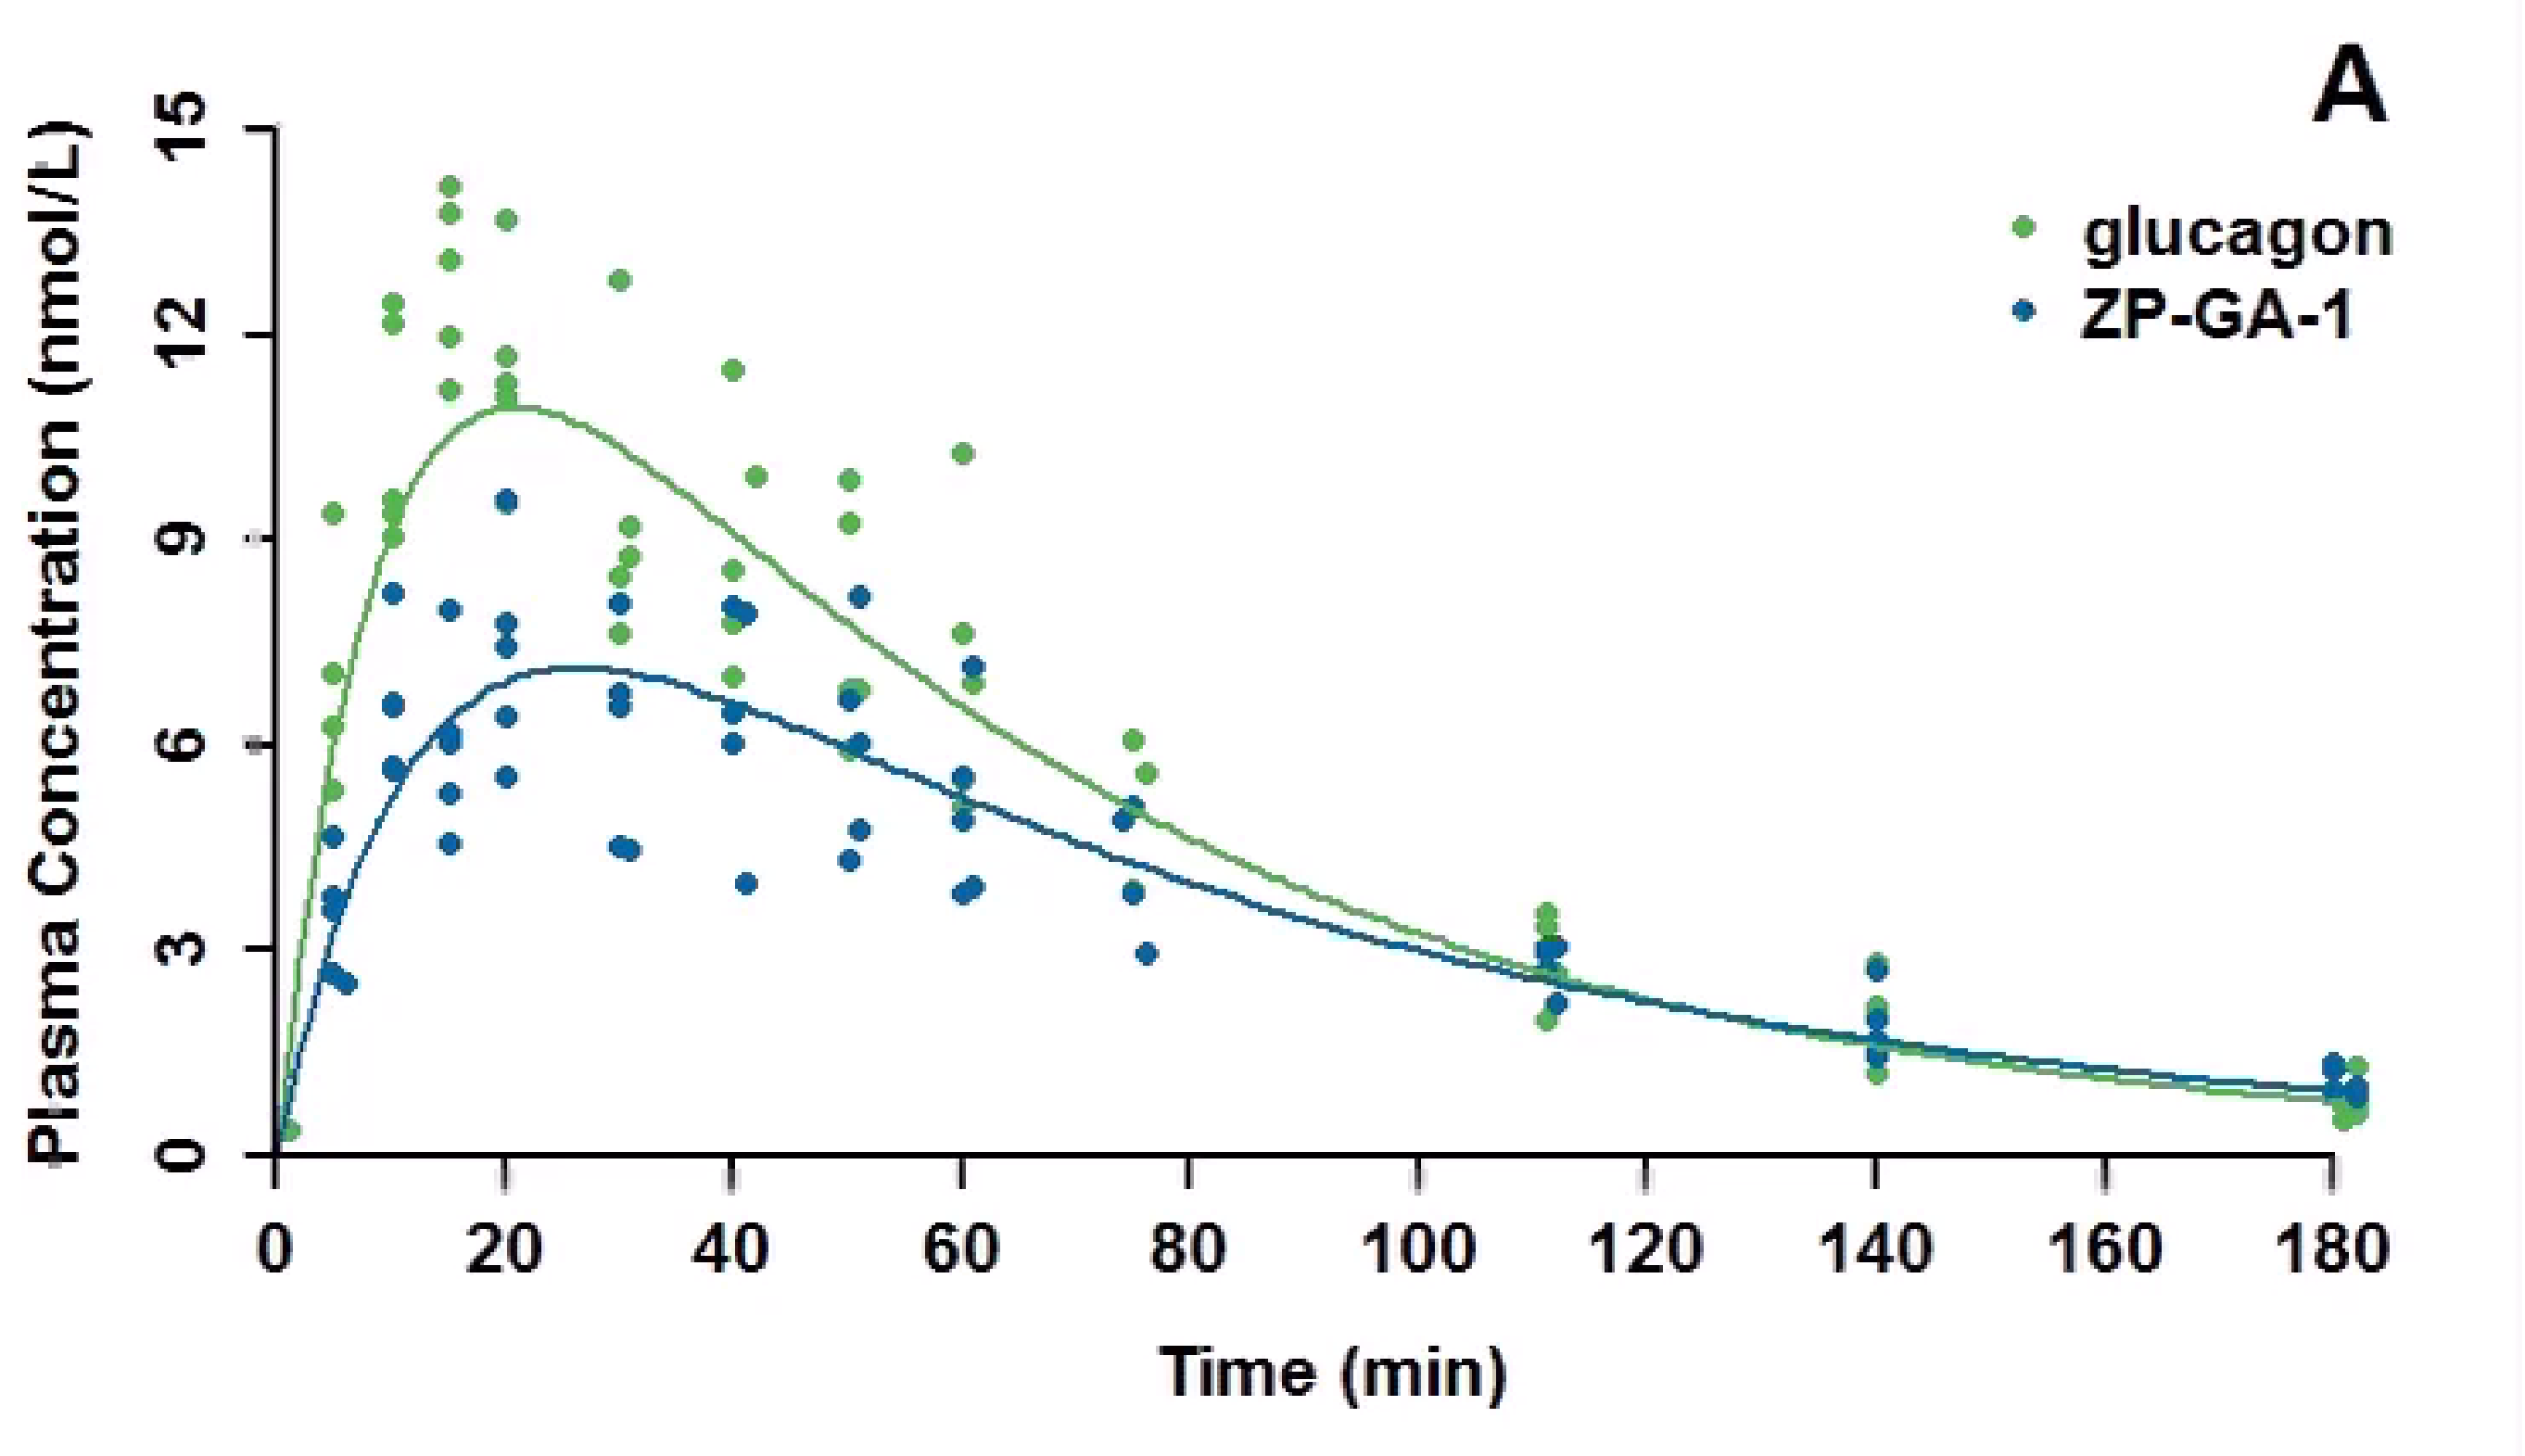
A
glucagon
ZP-GA-1
Plasma Concentration (nmol/L)
0
3
6
9
12
15
0
20
40
60
80
100
120
140
160
180
Time (min)

Figure 2: PK profiles after high SC dose. A) Raw observations with best fit to population data. B) Semi logarithmic plot with observations and best fit to population data. C) Semi logarithmic plot with individual fits.

	Glucagon (low SC dose)	**ZP-GA-1 (low SC dose)**	**Glucagon (high SC dose)**	**ZP-GA-1 (high SC dose)**
Variable	**Mean (±SD)**	**Mean (±SD)**	**Mean (±SD)**	**Mean (±SD)**
T_{max} (min)	24 (±14)	30 (±11)	20 (±5)	26 (±9)
C_{max}/D (kg/L)	0.065 (±0.032)	0.043 (±0.008)	0.096 (±0.015)	0.058 (±0.010)
k01 (min^{-1})	0.26 (±0.39)	0.07 (±0.05)	0.12 (±0.05)	0.11 (±0.10)
k10 (min^{-1})	0.016 (±0.007)	0.025 (±0.027)	0.018 (±0.004)	0.016 (±0.007)
V_F/W (L/kg)	12.7 (±3.9)	13.4 (±5.2)	7.4 (±1.5)	11.6 (±4.0)

Table 1: Population means and standard deviations (SD) of model parameters (k_{01}, k_{10}, V_F/W) and model fit characteristics (T_{max}, C_{max}/D).

Variable	**Dependency**	**P-value**
C_{max}/D (kg/L)	Compound	0.04

Table 2: Significant variable and its dependency according to a two-way (Dose*Compound) within subjects ANOVA analysis. P-values of remaining model parameters and model fit characteristics were not significant.

Conclusions

Zealand’s novel glucagon analogue, ZP-GA-1, shows similar PK characteristics (absorption, elimination, volume of distribution) to native glucagon. Time to maximum concentration, T_{max}, is also similar between native glucagon and ZP-GA-1. However, ZP-GA-1 has lower maximum concentration, C_{max}, than native glucagon after same level of dose administration.

Copyright © 2015 Sabrina Lyngbye Wendt slw@zealandpharma.com

ATTD, February 18-21 2015, Paris

REVOLUTIONARY HEALTH SOLUTIONS